Revealing the geometry of duration space

Camille L. Grasso[1,2,3], Ladislas Nalborczyk[4], Virginie van Wassenhove[1]

[1] Cognitive Neuroimaging Unit (UNICOG), Université Paris-Saclay, CEA, Inserm, NeuroSpin, Gif-sur-Yvette, France
[2] Department of Translational Neuroscience, Wake Forest University School of Medicine, Winston-Salem, NC, USA
[3] Univ-Grenoble Alpes, Univ. Savoie Mont Blanc, CNRS, LPNC, Grenoble, France
[4] Aix Marseille Univ, CNRS, LPL, Aix-en-Provence, France

camille.grasso@univ-grenoble-alpes.fr

Is duration represented as a 1D mental timeline, or as a richer geometrical structure?

Several influential frameworks propose that durations are magnitudes organised along a mental timeline: a single internal dimension on which shorter and longer intervals are ordered monotonically [1–4]. Here, we asked whether such a one-dimensional account is sufficient to characterise the psychological and neural representations of duration; **or whether they are better described by a richer geometry**. We addressed this question in 33 participants by combining pairwise similarity judgements with EEG recorded during an oddball task using the same auditory durations, and by comparing behavioural and time-resolved neural RDMs against theoretical models of duration organisation [5-6]. This approach allowed us to investigate both the latent geometry of subjective duration space and its temporal expression in neural activity.

Experimental protocols

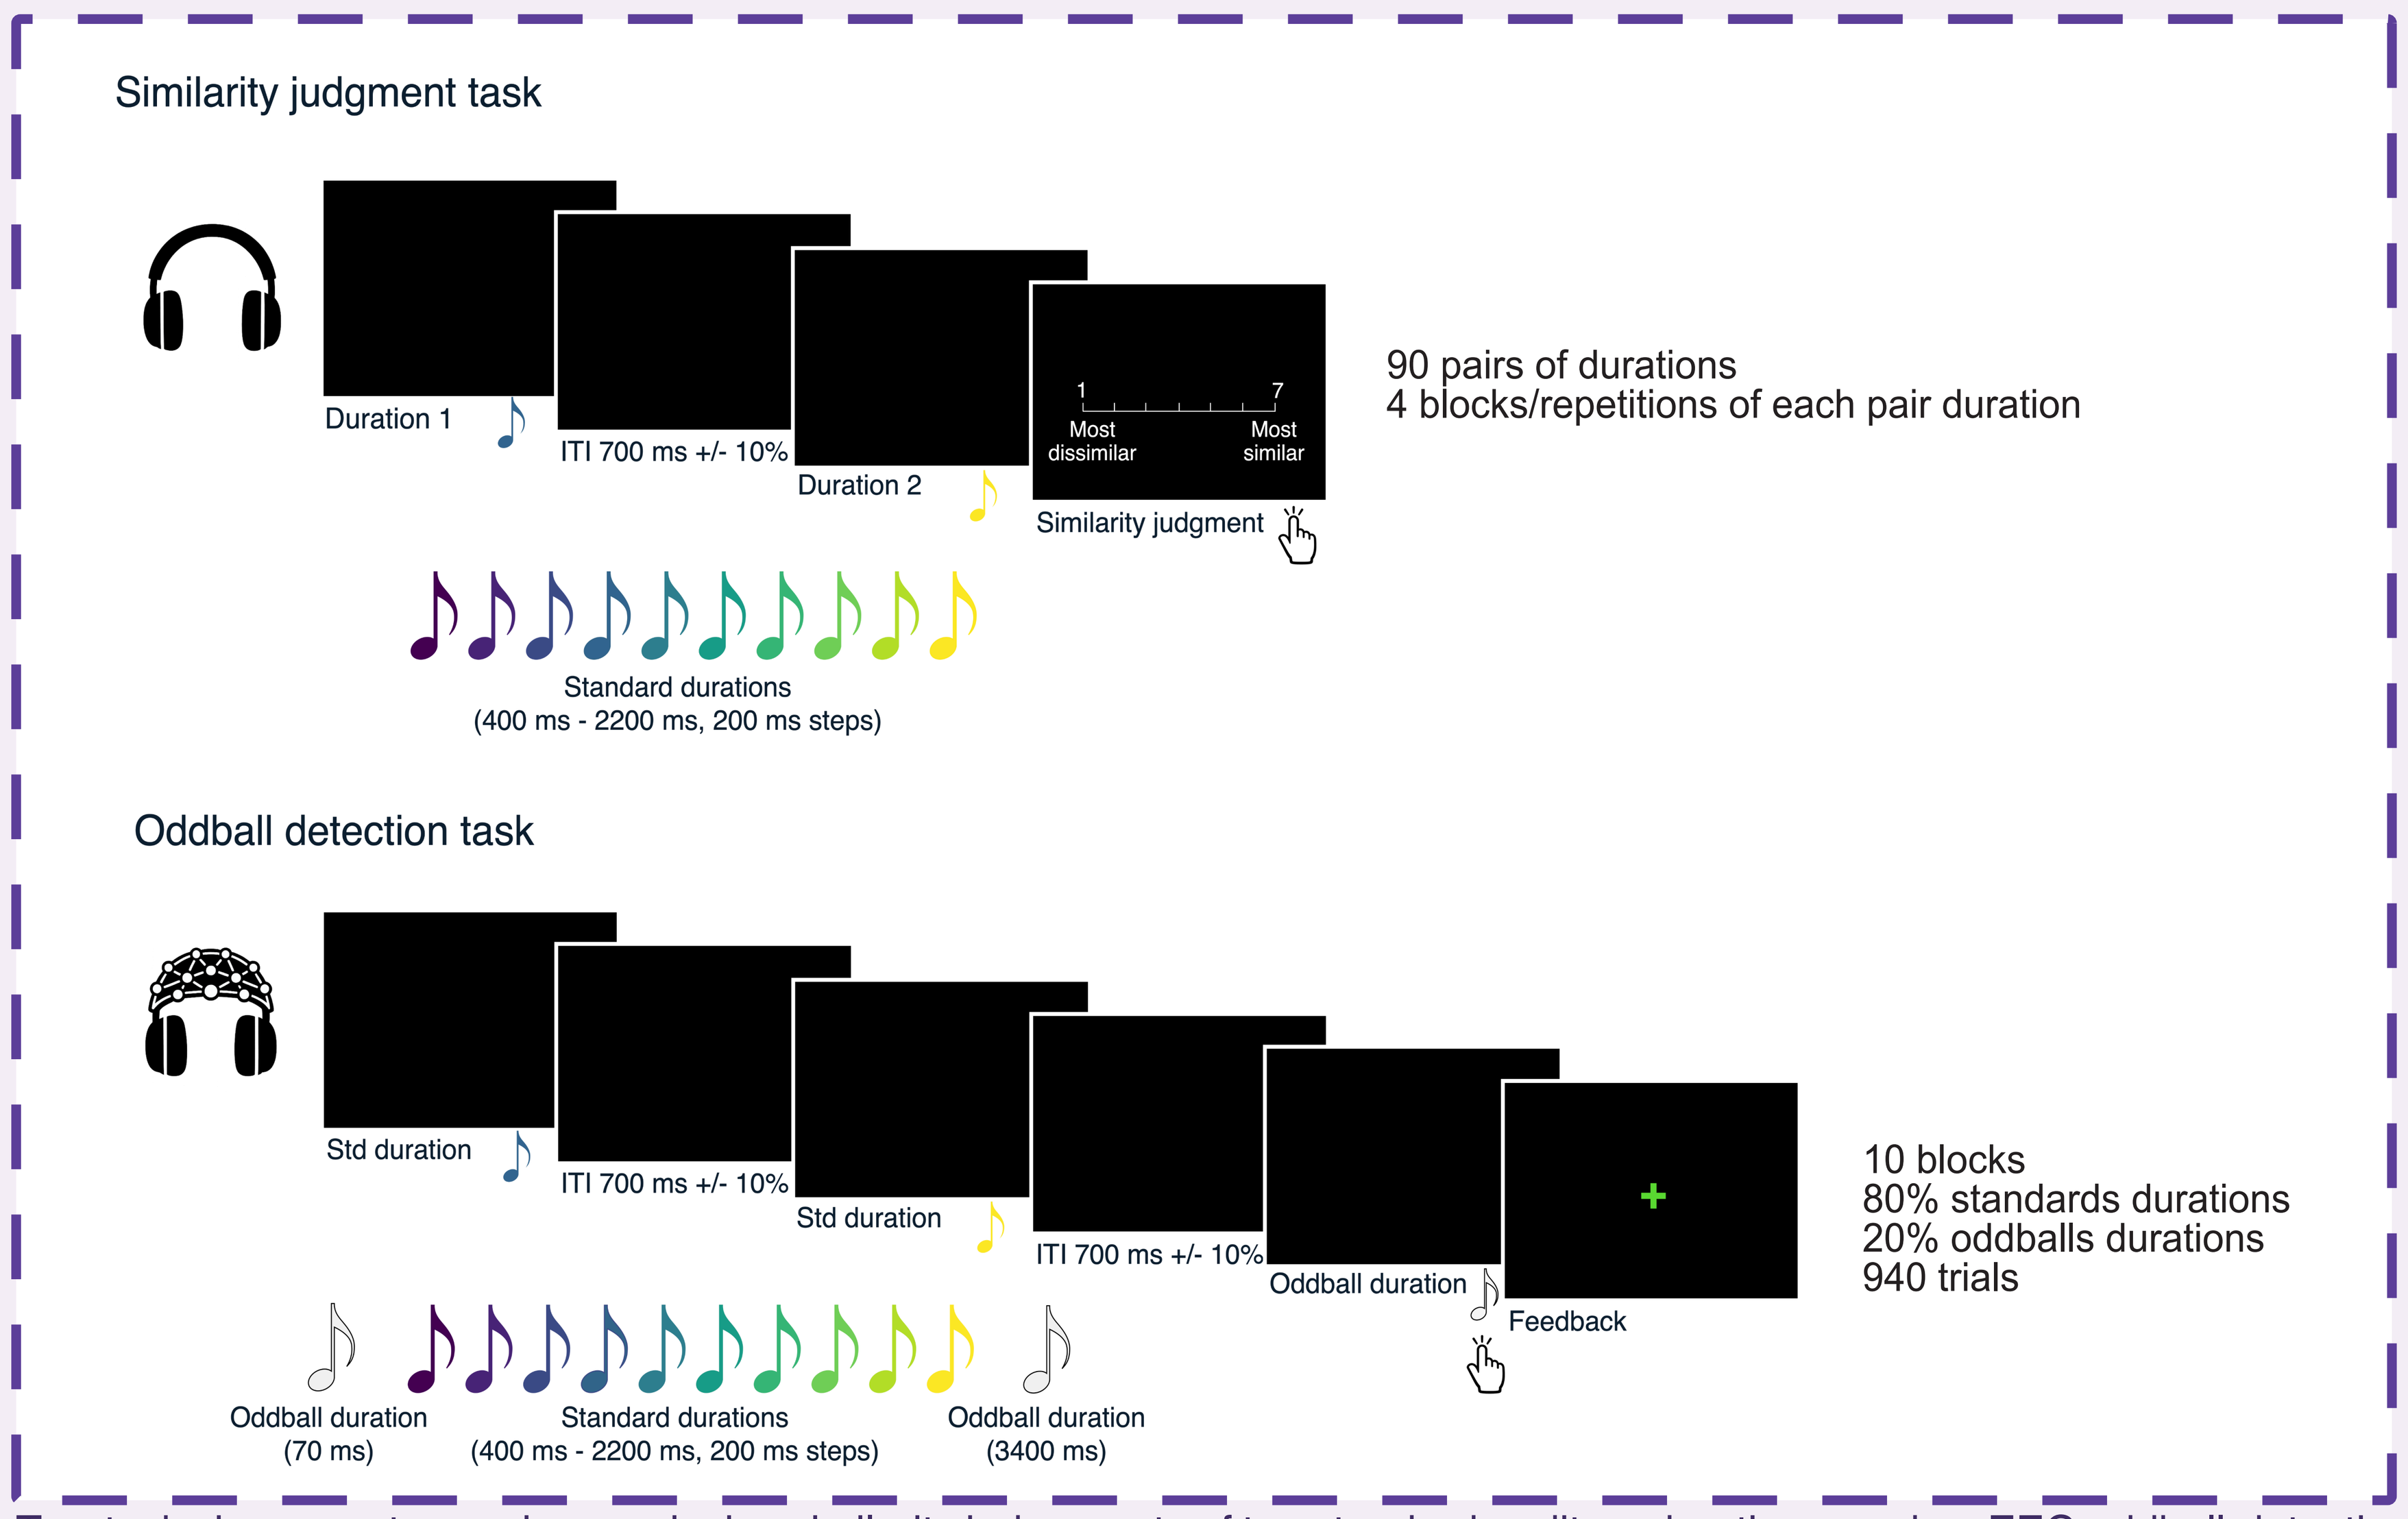
Similarity judgment task
Duration 1
ITI 700 ms +/- 10%
Duration 2
1
7
Most dissimilar
Most similar
Similarity judgment
90 pairs of durations
4 blocks/repetitions of each pair duration
Standard durations
(400 ms - 2200 ms, 200 ms steps)
Oddball detection task
Std duration
ITI 700 ms +/- 10%
Std duration
ITI 700 ms +/- 10%
Oddball duration
Feedback
10 blocks
80% standards durations
20% oddballs durations
940 trials
Oddball duration
(70 ms)
Standard durations
(400 ms - 2200 ms, 200 ms steps)
Oddball duration
(3400 ms)

Two tasks in separate sessions: pairwise similarity judgements of ten standard auditory durations, and an EEG oddball-detection task using the same standards. Resulting behavioural and time-resolved EEG RDMs were compared with six theoretical RDMs: Linear, Logarithmic, Binary, Power, Helix, Random (control), plus a task-specific Regularness RDM encoding each standard duration's distance to the nearest oddball.

Behavioural geometry

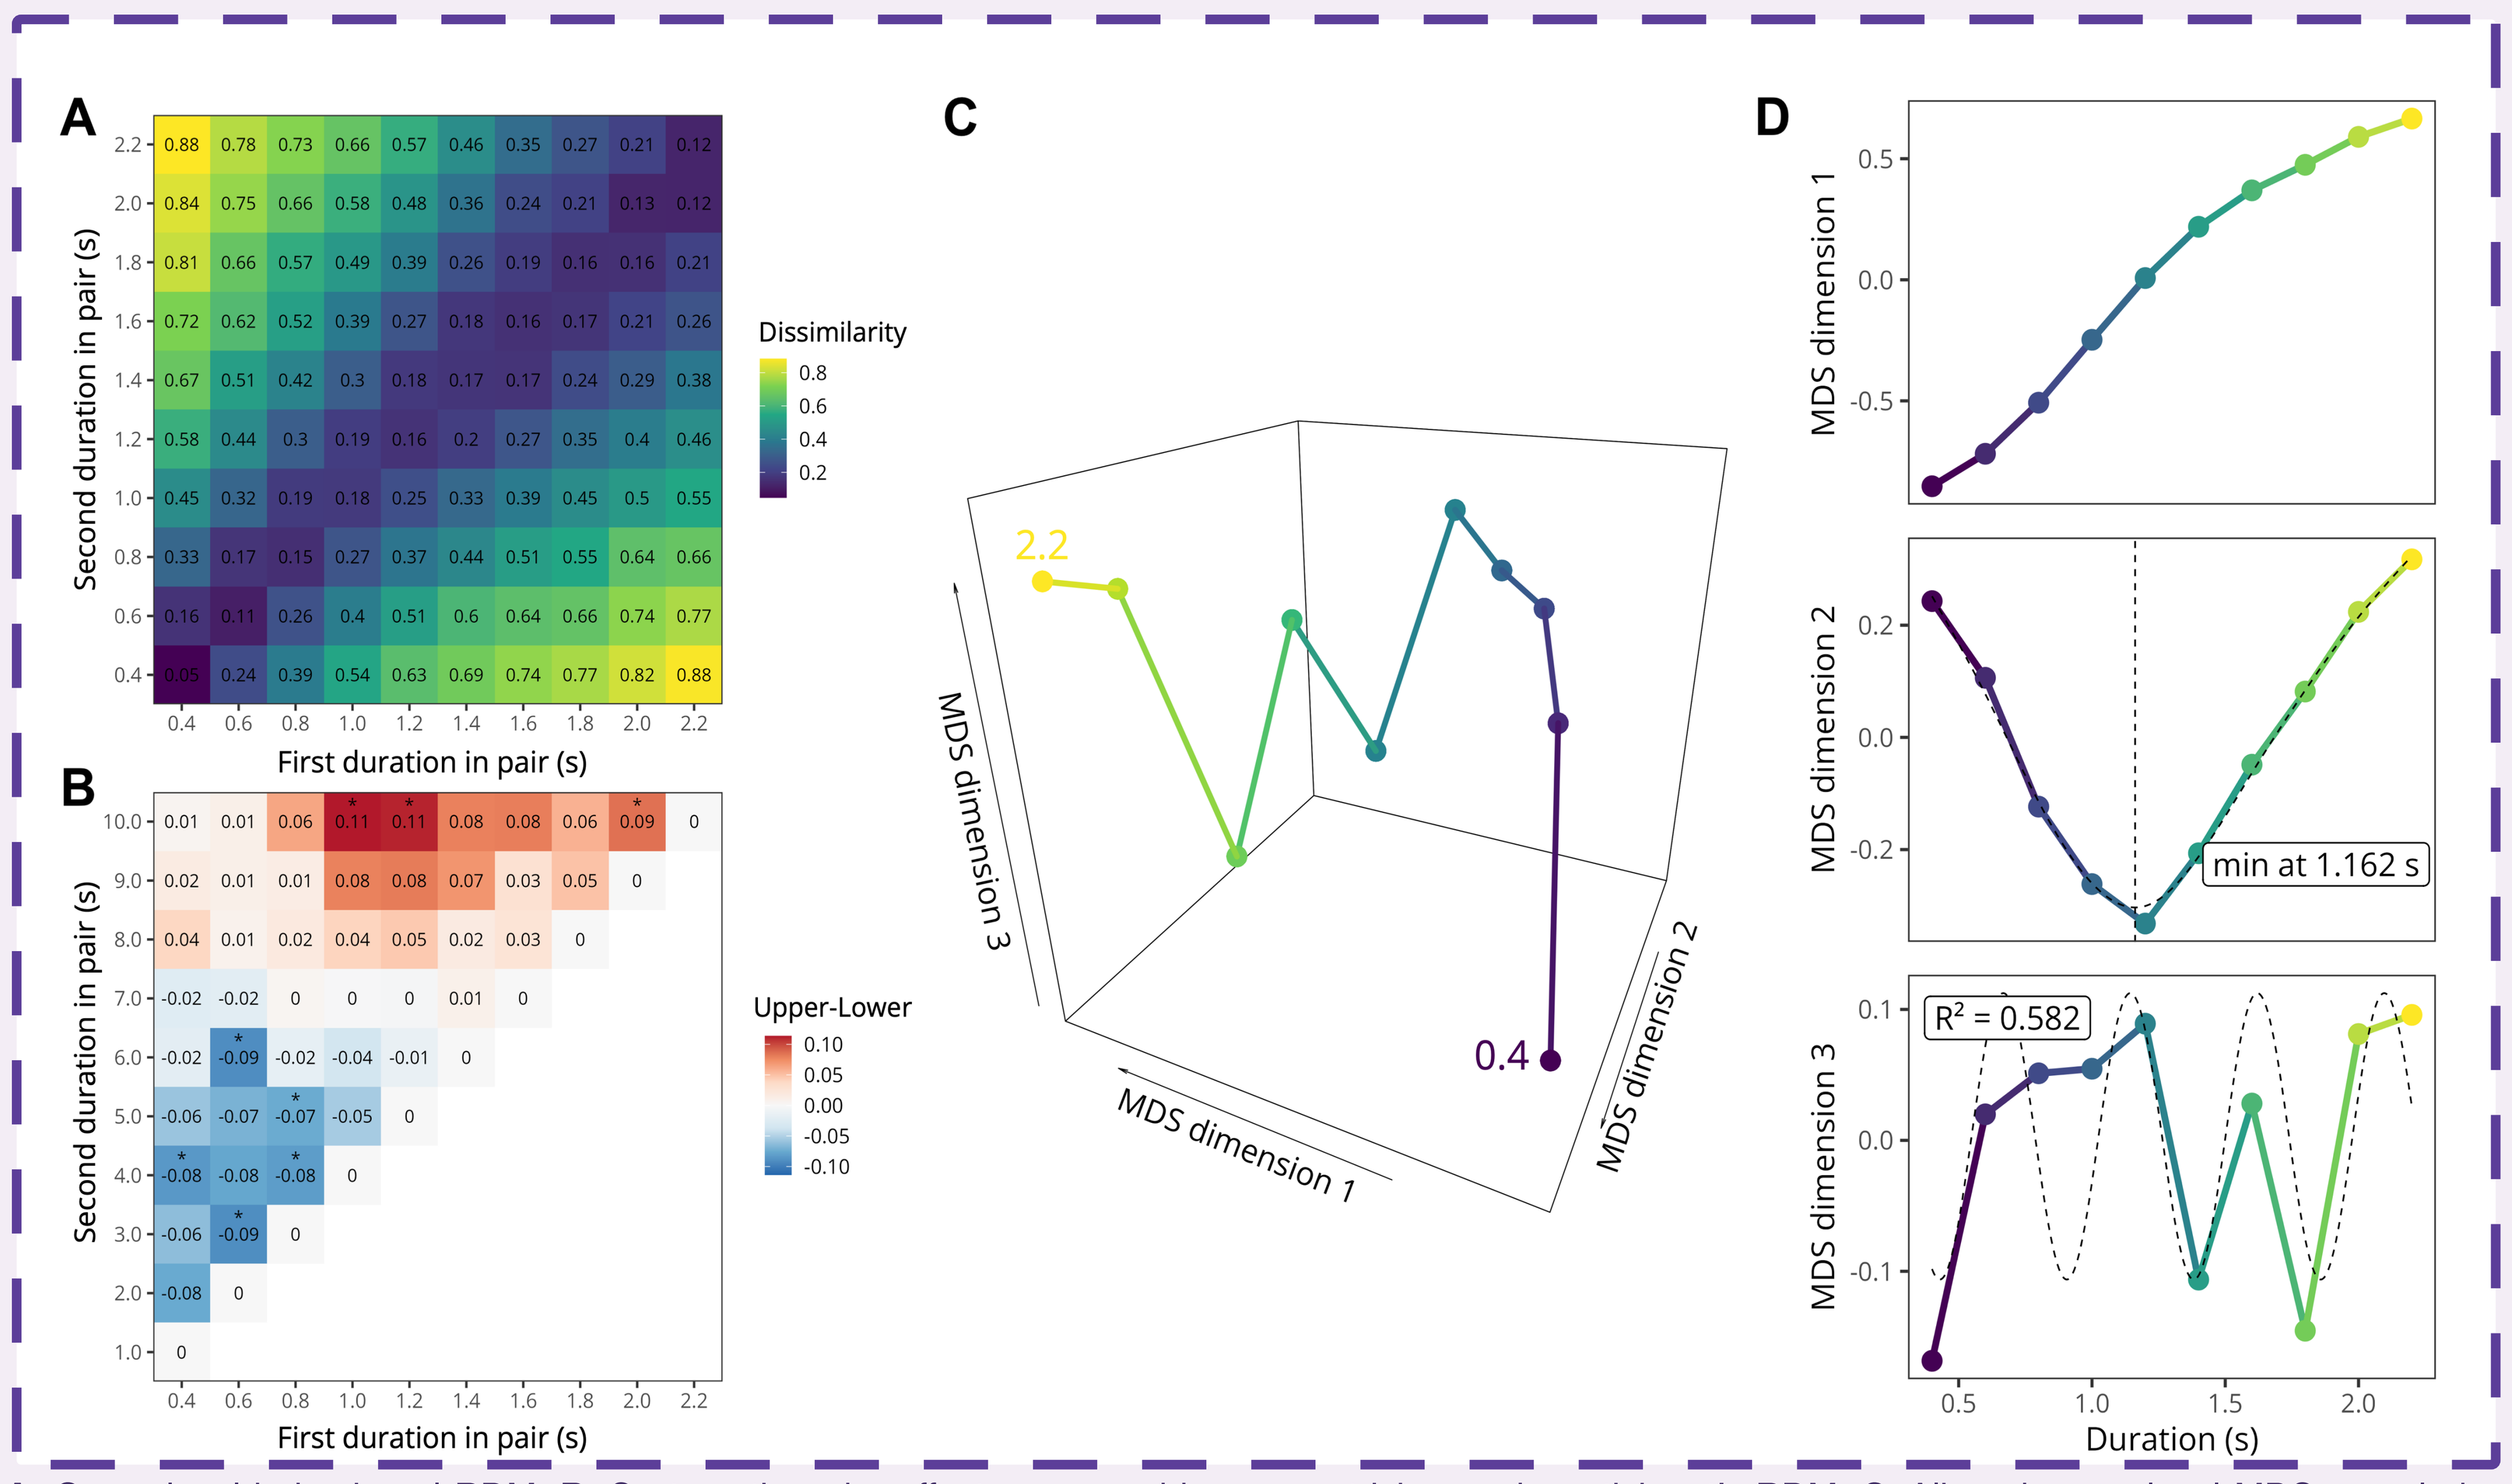
A
Second duration in pair (s)
First duration in pair (s)
Dissimilarity
B
Upper-Lower
C
2.2
0.4
MDS dimension 1
MDS dimension 2
MDS dimension 3
D
MDS dimension 1
MDS dimension 2
min at 1.162 s
MDS dimension 3
R² = 0.582
Duration (s)

A. Group-level behavioural RDM. **B.** Systematic order effects, removed by symmetrising each participant's RDM. **C.** Aligned group-level MDS revealed a spring-like geometry; three dimensions were retained because two complementary elbow criteria applied to the group-average stress curve both selected a 3D solution. **D.** Dimensions captured a dominant magnitude axis, a U-shaped distance-to-mean component, and a weaker periodic-like modulation.

Helix shape and endogenous resting-state oscillations

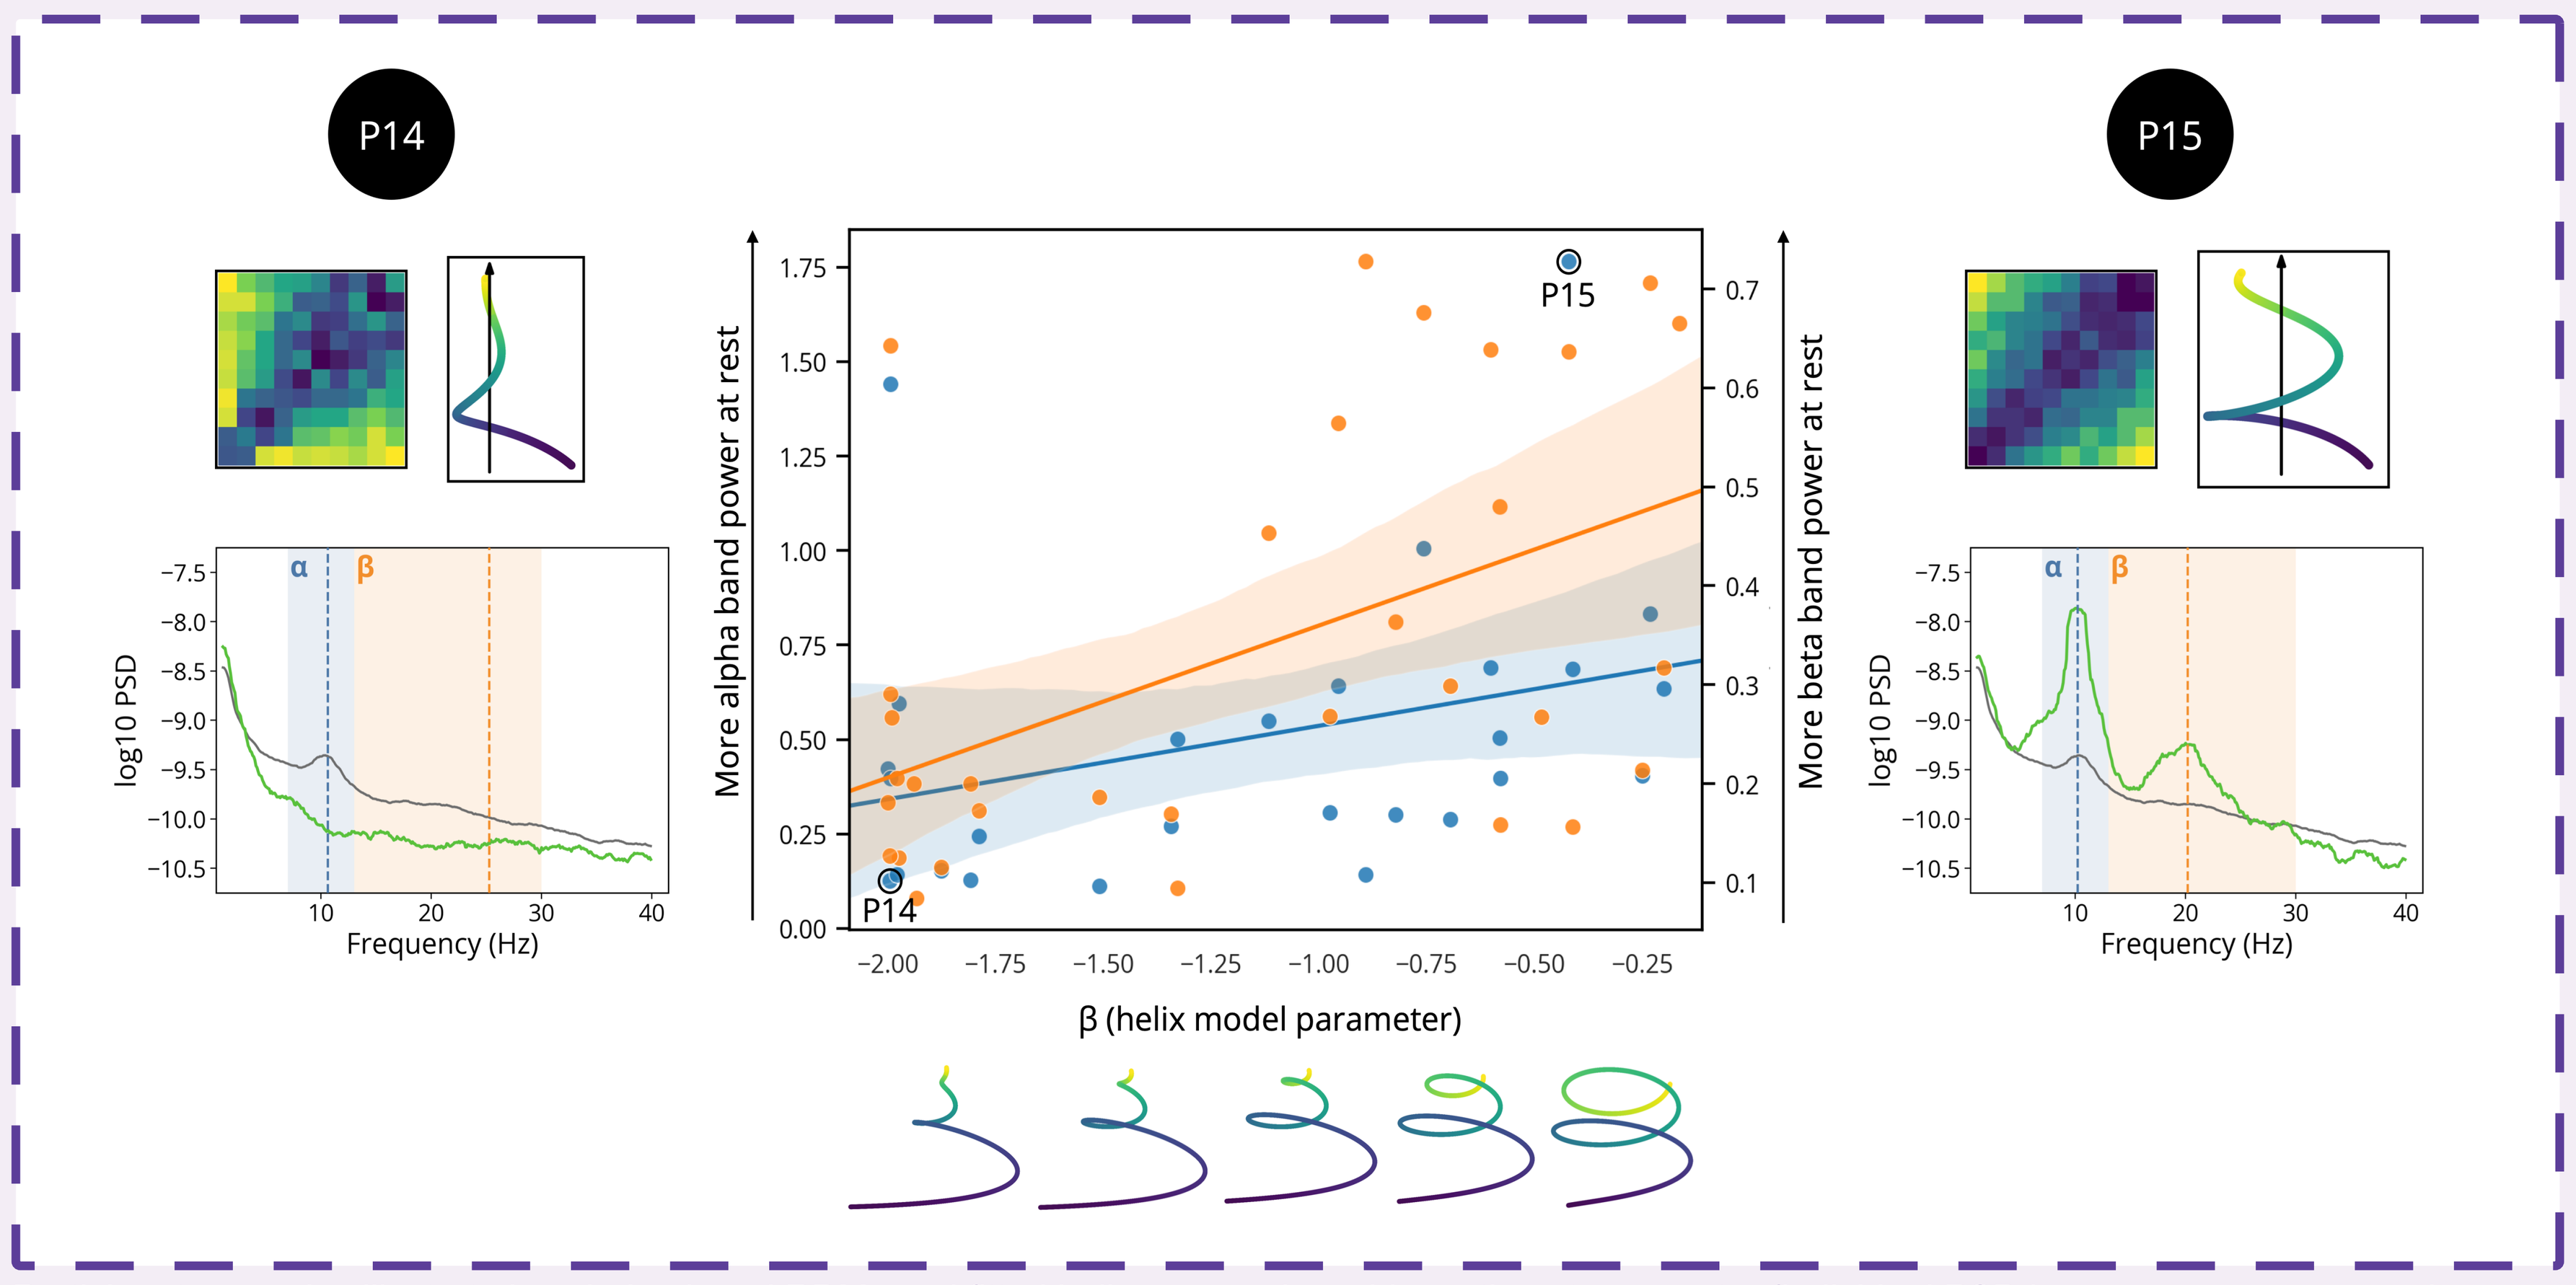
P14
P15
α
β
log10 PSD
Frequency (Hz)
More alpha band power at rest
More beta band power at rest
β (helix model parameter)

Participant examples illustrate how behavioural RDMs and fitted helices vary alongside resting-state spectra (individual PSD in green; group average in grey; alpha and beta bands in blue and orange). Across participants, resting alpha and beta power measured in a separate session correlated with the behavioural helix β parameter—values closer to zero indicating a less compressed, more spring-like geometry, but not with the Power-law exponent.

[1] Bonato et al. (2012). Neurosci Biobehav Rev. [2] Gibbon (1977). Psychol Rev. [3] Gallistel & Gibbon (2000). Psychol Rev.
[4] Stevens (1957). Psychol Rev. [5] Shepard & Chipman (1970). Cogn Psychol. [6] Kriegeskorte & Kievit (2013). Trends Cogn Sci.

Neural geometry

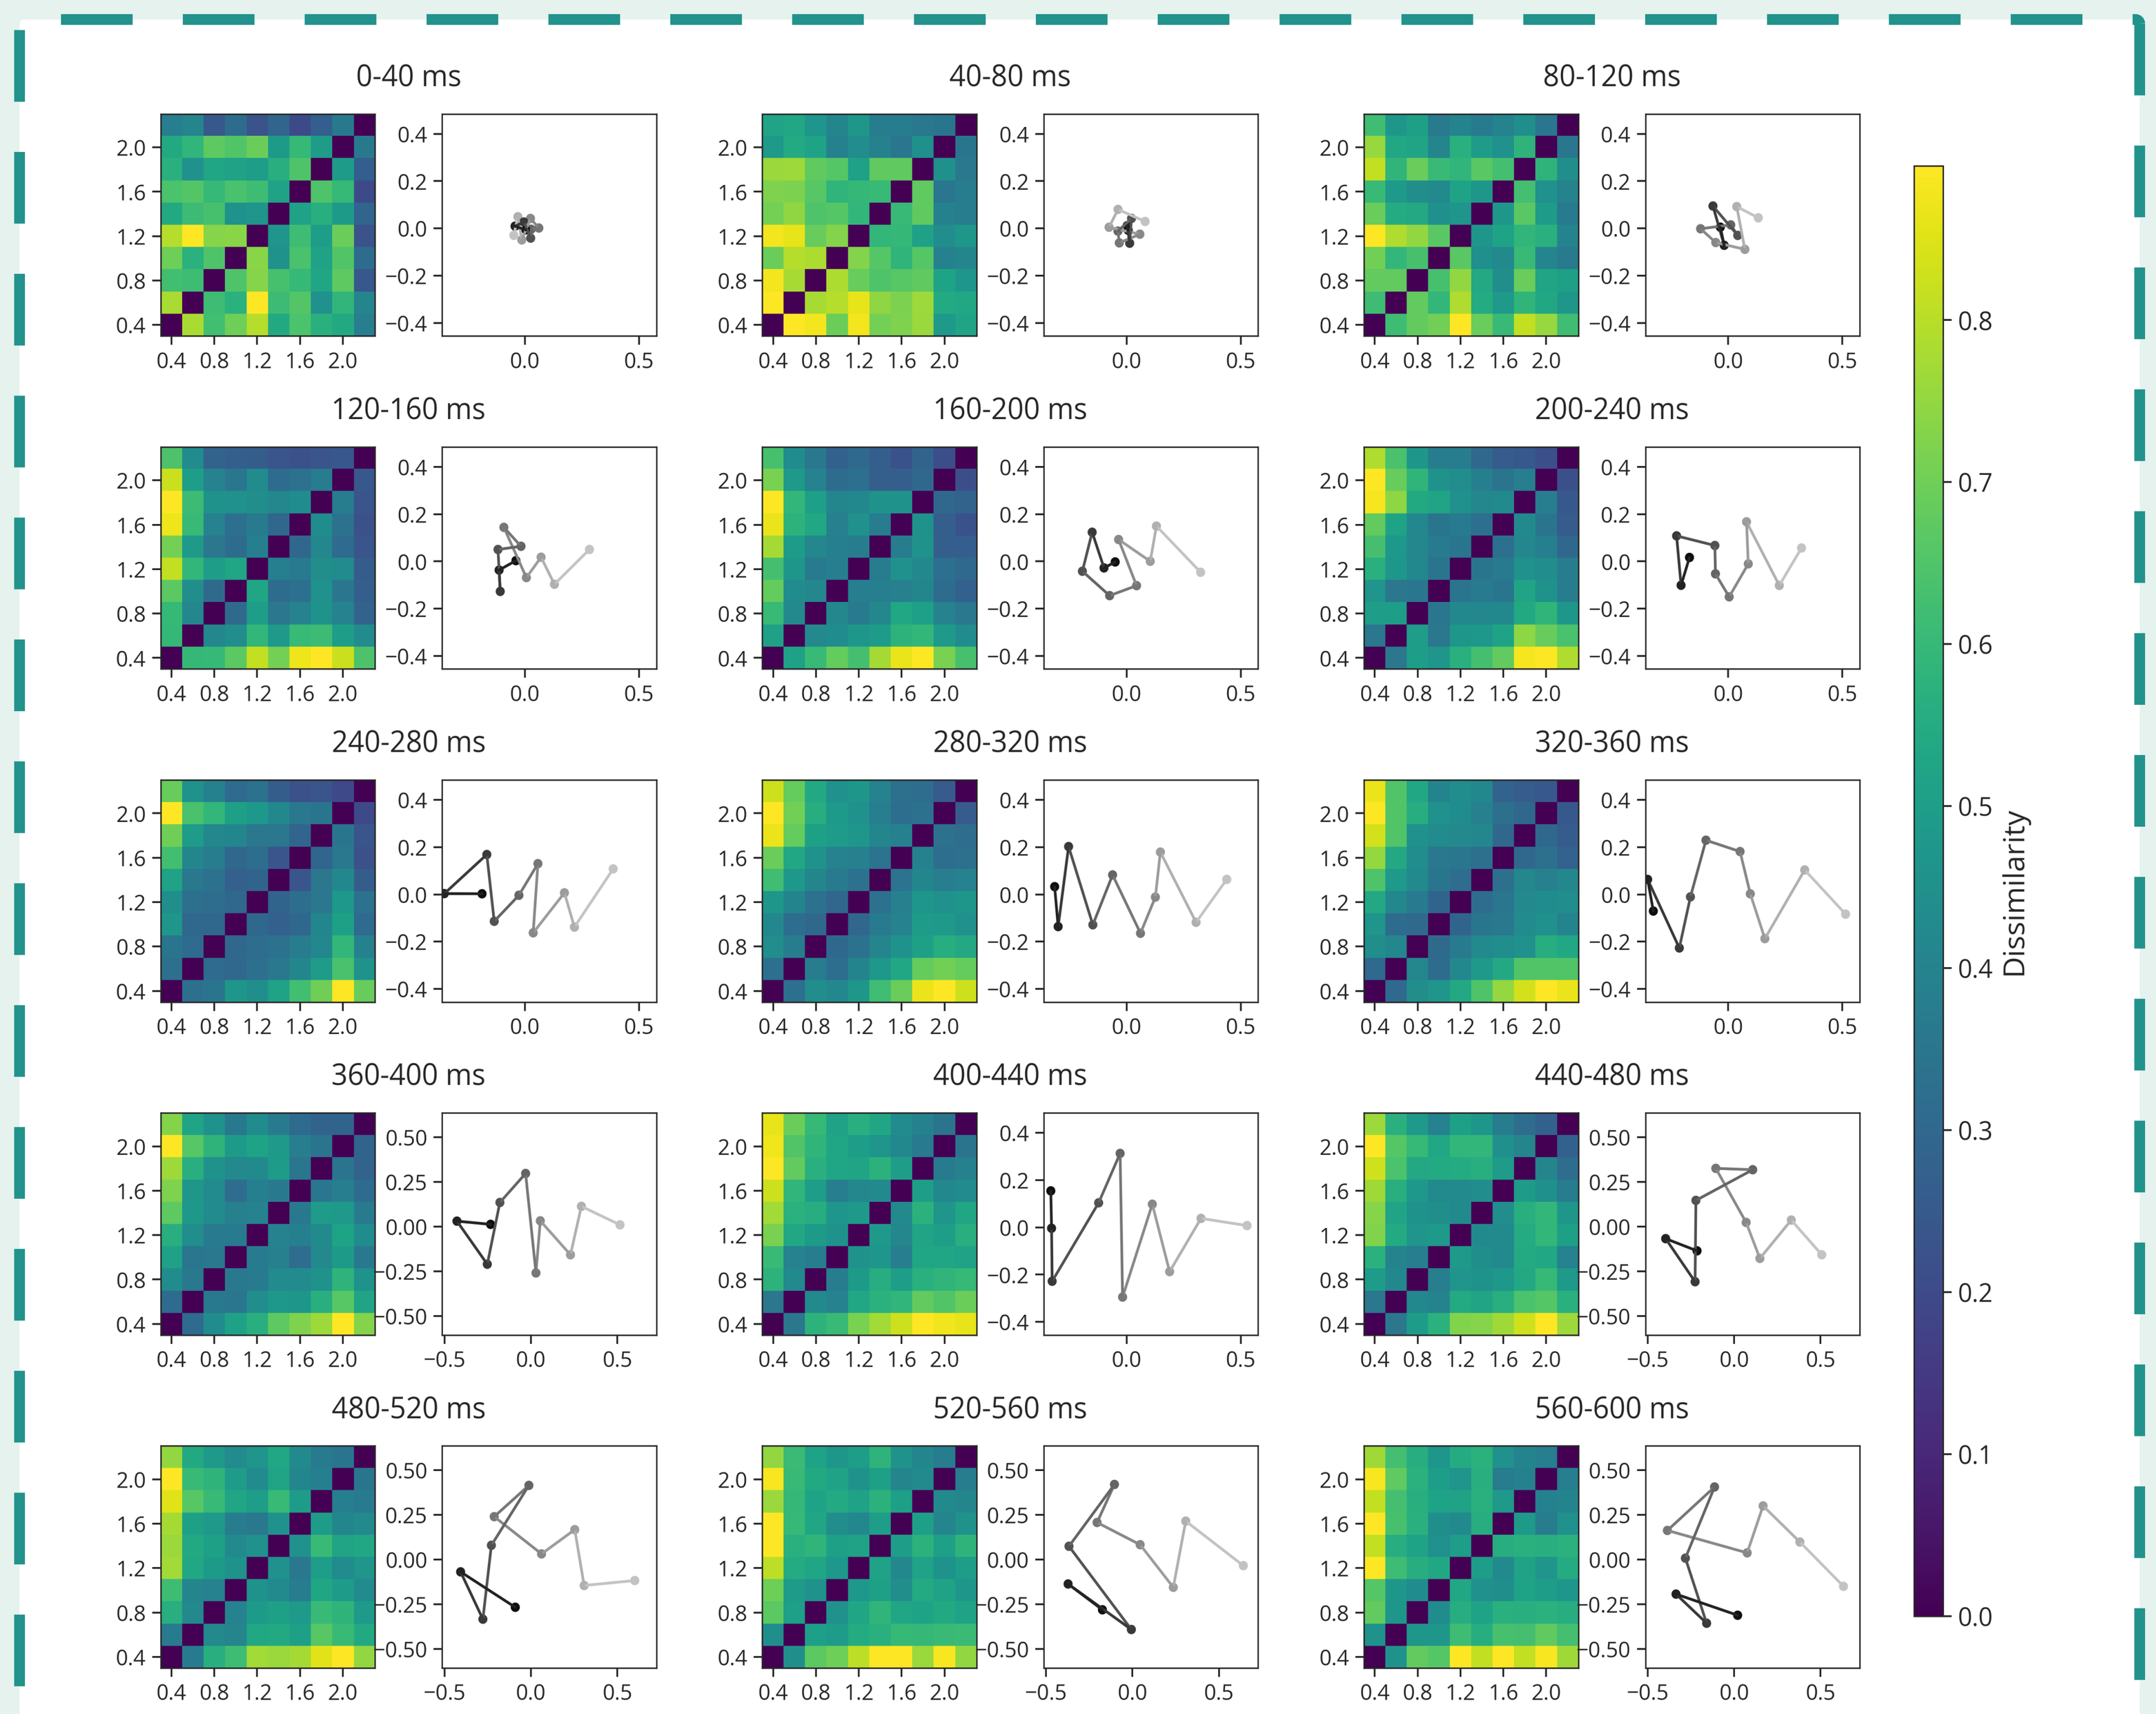
0-40 ms
40-80 ms
80-120 ms
120-160 ms
160-200 ms
200-240 ms
240-280 ms
280-320 ms
320-360 ms
360-400 ms
400-440 ms
440-480 ms
480-520 ms
520-560 ms
560-600 ms
Dissimilarity

For each 40-ms window after duration offset, group-average EEG RDMs and corresponding 2D MDS projections track the evolving neural geometry of duration. Early RDMs were weakly structured and MDS points remained clustered; from ~160–200 ms, durations became increasingly differentiated and systematically organised. Around 300 ms, this geometry shifted toward the structure observed in behavioural similarity judgements.

Representational Similarity Analysis

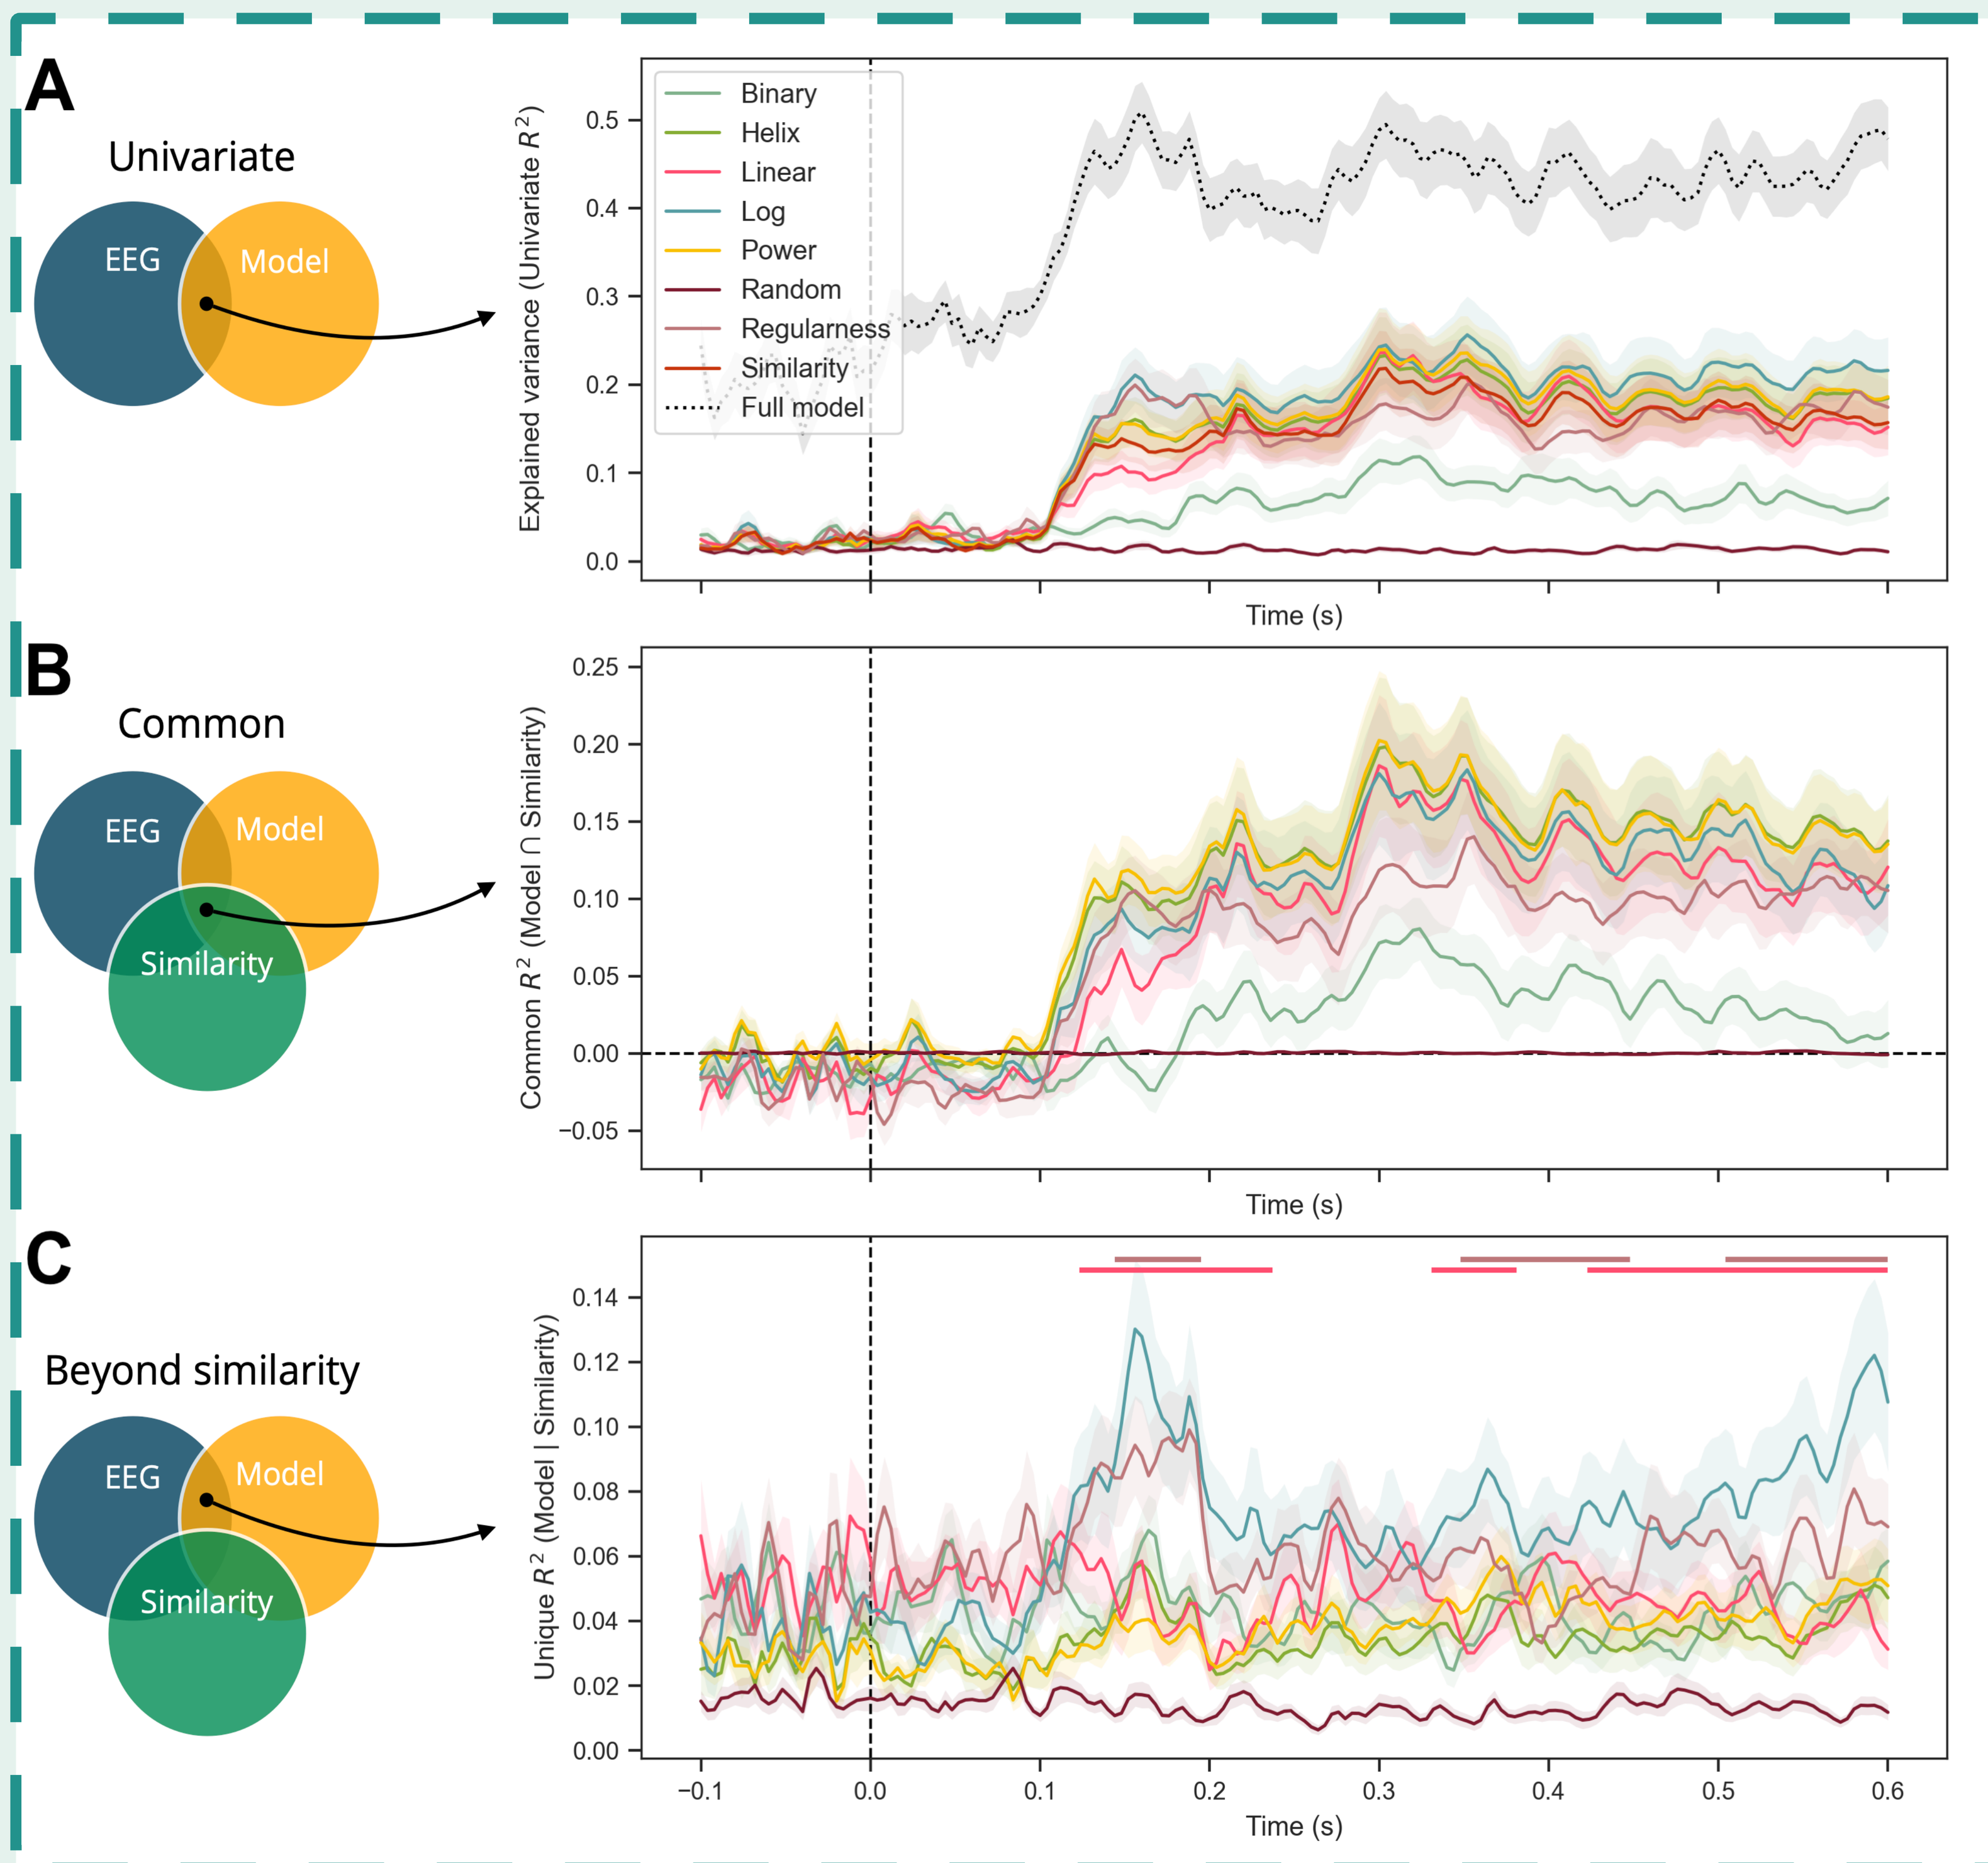
A
Univariate
EEG
Model
Explained variance (Univariate R^2)
Binary
Helix
Linear
Log
Power
Random
Regularness
Similarity
Full model
Time (s)
B
Common
EEG
Model
Similarity
Common R^2 (Model ∩ Similarity)
Time (s)
C
Beyond similarity
EEG
Model
Similarity
Unique R^2 (Model | Similarity)
Time (s)

Group-level time-resolved RSA and commonality analyses. **A.** Variance in EEG RDMs explained by each theoretical model and by the full model, which accounted for ~40–50% of the variance with peaks around 150 and 300 ms after duration offset. **B.** Shared variance between each model, EEG, and behavioural similarity increased after ~100 ms and peaked around 300 ms, indicating convergence between neural and behavioural geometries. **C.** Beyond behavioural similarity, Logarithmic and Regularness models captured additional early neural structure, particularly around 150–200 ms. Horizontal bars indicate periods in which the Logarithmic model outperformed Regularness or Linear models (posterior odds > 20).

CONCLUSIONS Results revealed a multidimensional organisation of duration space, with a dominant magnitude dimension. However, this axis alone did not capture the full geometry. A second dimension reflected distance to the geometric mean of the stimulus set (not reducible to a simple MDS horseshoe effect) suggesting that durations are encoded relationally with respect to their temporal context, rather than solely as absolute magnitudes. A third, weaker dimension showed a periodic-like modulation, consistent with a recurrent structure. EEG analyses showed that the neural geometry unfolded in successive stages: a first early stage after duration offset, in which EEG RDMs are primarily reflected by a logarithmic compression and task-relevant structure, and a later stage, around 300 ms, in which the geometry became more aligned with behavioural RDMs and with the Helix and Power models that best captured it. Finally, resting-state alpha and beta power correlated specifically with the helix β parameter fitted to behavioural similarity judgements (measured in separate sessions), but not with the Power-law exponent: stronger oscillatory power was associated with a less compressed, more spring-like duration space. Together, these findings move beyond a purely one-dimensional mental timeline toward a multidimensional and dynamic duration geometry.